In Vivo Efficacy of Novel Sodium Glucose Co-transporter 2 (SGLT2) Inhibitor SBM-TFC-039 on Glucose Excretion and Tolerance in Normal Sprague-Dawley and Zucker Diabetic Fatty Rats.

Bertrand Plouvier*, Marie-Claude Roy†, Elena-Dana Baraboi†, Denis Richard†
*Sirona Biochem Corp., Vancouver, BC, CANADA, info@sironabiochem.com
†Centre de recherche de l'IUCPQ, Québec, QC, CANADA

Abstract

Sirona Biochem and TFChem have developed a new sodium glucose co-transporter 2 (SGLT2) inhibitor, SBM-TFC-039, for the treatment of diabetes. SGLT2 inhibitors constitute a new class of oral antidiabetic agents that target the sodium glucose co-transporter. The co-transporter SGLT2 is responsible for the re-absorption of 90% of the glucose in the kidney. This new agent would improve glycaemic control by inducing glucosuria without any direct effect on insulin secretion.

Sirona Biochem and Institut Universitaire de Cardiologie et de Pneumologie de Québec (IUCPQ) assessed the urinary glucose excretion (UGE) and response to glucose challenges following oral administration in normal Sprague Dawley (SD) and Zucker diabetic fatty (ZDF) rats. The chronic administration in ZDF rats for 28 days was also investigated.

SBM-TFC-039 triggered glucosuria in a dose dependent manner following acute oral administration to normal SD and diabetic ZDF rats. Also, in SD and ZDF rats, SBM-TFC-039 significantly reduced blood glucose excursions following a glucose challenge at 1 hour after drug administration. Although less apparent, the glucose tolerance improving effect of SBM-TFC-039 was still significant when the glucose challenge was performed 18 hours after oral administration. In ZDF rats treated with SBM-TFC-039, glycaemia decreasing rates and UGEs were strongly correlated. When chronically administered (for 28 days) in ZDF rats, SBM-TFC-039 significantly reduced ad libitum and fasting glycaemia at 2 weeks after the beginning of the treatment.

SBM-TFC-039 also reduced fructosamine and fructosamine:total protein ratio after 4 weeks of treatment. Those results demonstrate the ability of SBM-TFC-039 to improve glucose metabolism in normal rats and to lower the glycaemia in ZDF diabetic rats.

Methods

For all animal studies, the vehicle used for drug administration was 5% 1-methyl-2-pyrrolidinone, 20% polyethyleneglycol, and 20 mmol/L sodium diphosphate solution (v/v/v).

Urinary glucose excretion (UGE) and glucose tolerance in normal SD rats. SD rats were placed into metabolic cages for baseline urine collection over 24 hours. After a period of 18 hours of acclimatization in the metabolic cages, rats were weighed, randomized (n = 6) and dosed orally with single doses of vehicle (10 mL/kg) or SBM-TFC-039 (1, 3 or 10 mg/kg). Immediately after dosing, rats were returned to the metabolic cages for urine collection. Urine was collected and weighted at time 0, 2, 4, 6, 22 and 24 hours after oral gavage. They had free access to food and water. After urine volume was measured, the urinary glucose concentration (UGC) was determined using a Wako glucose analyzer kit (Wako Diagnostics, Richmond, VA, USA). UGE was calculated by multiplying UGC by urinary volumes. After one week recovery the same rats were fasted overnight (for 18 hours), weighed, and randomized (n = 6). SBM-TFC-039, at 1 mg/kg, and the vehicle, at 10 mL/kg, were administered orally 1 hour before the oral glucose tolerance test (OGTT). The OGTT consisted of the administration of a glucose solution (20%, 2 g/kg), given orally. Blood was obtained (puncture of the saphenous vein) at 0, 5, 15, 30, 60 and 120 minutes. Blood glucose was measured using a glucometer (One Touch, Johnson & Johnson).

Glycaemia and fructosamine level in ZDF rats. Male obese ZDF rats (strain 370, 6 per group), aged 13 weeks, and 6 male lean fa/+ rats (strain 380), aged 13 weeks were used. Rats were weighed and given vehicle by oral gavage vehicle (10 mL/kg) or SBM-TFC-039 (1 mg/kg). They had free access to food and water. Blood samples were collected (puncture of the saphenous vein) immediately before dosing (time 0) and at 2, 6, and 24 hours after drug administration. SBM-TFC-039 (1 mg/kg) or vehicle (10 mL/kg) was then orally administered once daily for 28 days. Fed blood glucose was measured once a week (on days 0, 7, 14, 21 and 28).

Data analysis. The results were expressed as means ± SEMs. Statistical differences between groups were determined using one- or two-way ANOVA followed by Bonferroni's or Fisher multiple comparisons. Body weight, food intake and water intake results from the chronic experiment were analyzed using repeated measures ANOVA. The correlations between UGE and blood glucose levels were estimated using Pearson's correlation analysis. Statistical analyses were performed using Graphpad Prism. The data were tabulated in Excel and plotted in Graphpad Prism.

Results

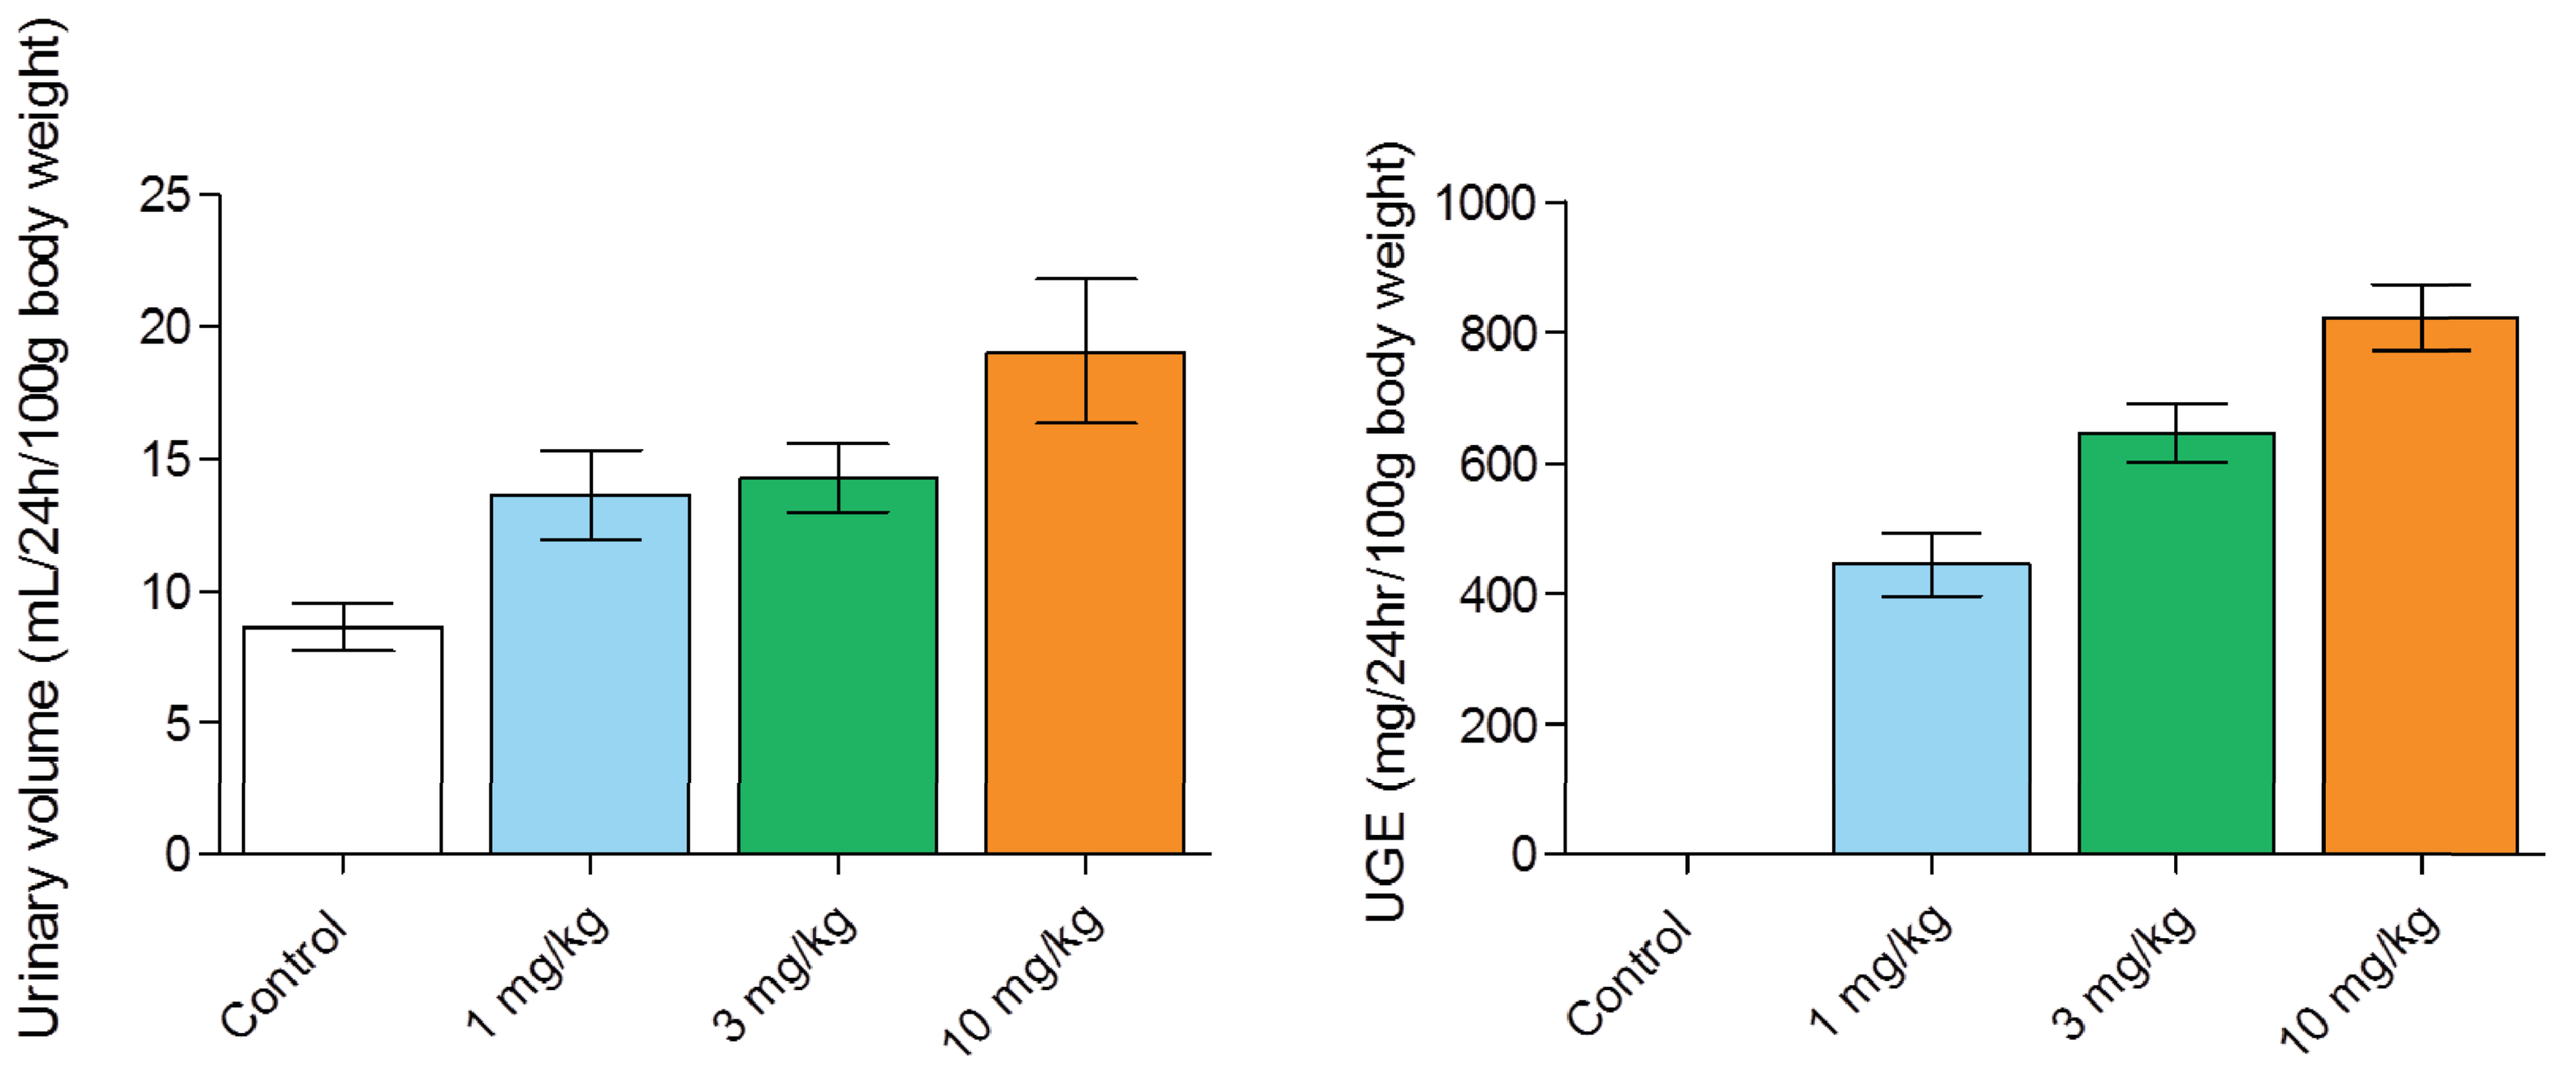
Urinary volume (mL/24h/100g body weight)
UGE (mg/24hr/100g body weight)
Control
1 mg/kg
3 mg/kg
10 mg/kg

Figure 1: Urinary volume and urinary glucose excretion (UGE) per 100 g body weight

Figure 1 illustrates the ability of SBM-TFC-039 to increase urine volume and glucose excretion in a dose dependent manner in SD rats. Results in Figure 2 demonstrate the ability of SBM-TFC-039 to improve glucose tolerance. The improvement was particularly apparent when the OGTT was performed 1 hour after the administration of the drug candidate (Figure 2, left panel). SBM-TFC-039 at 1 mg/kg po decreased blood glucose excursions by 34% following a glucose challenge (Figure 2, right panel).

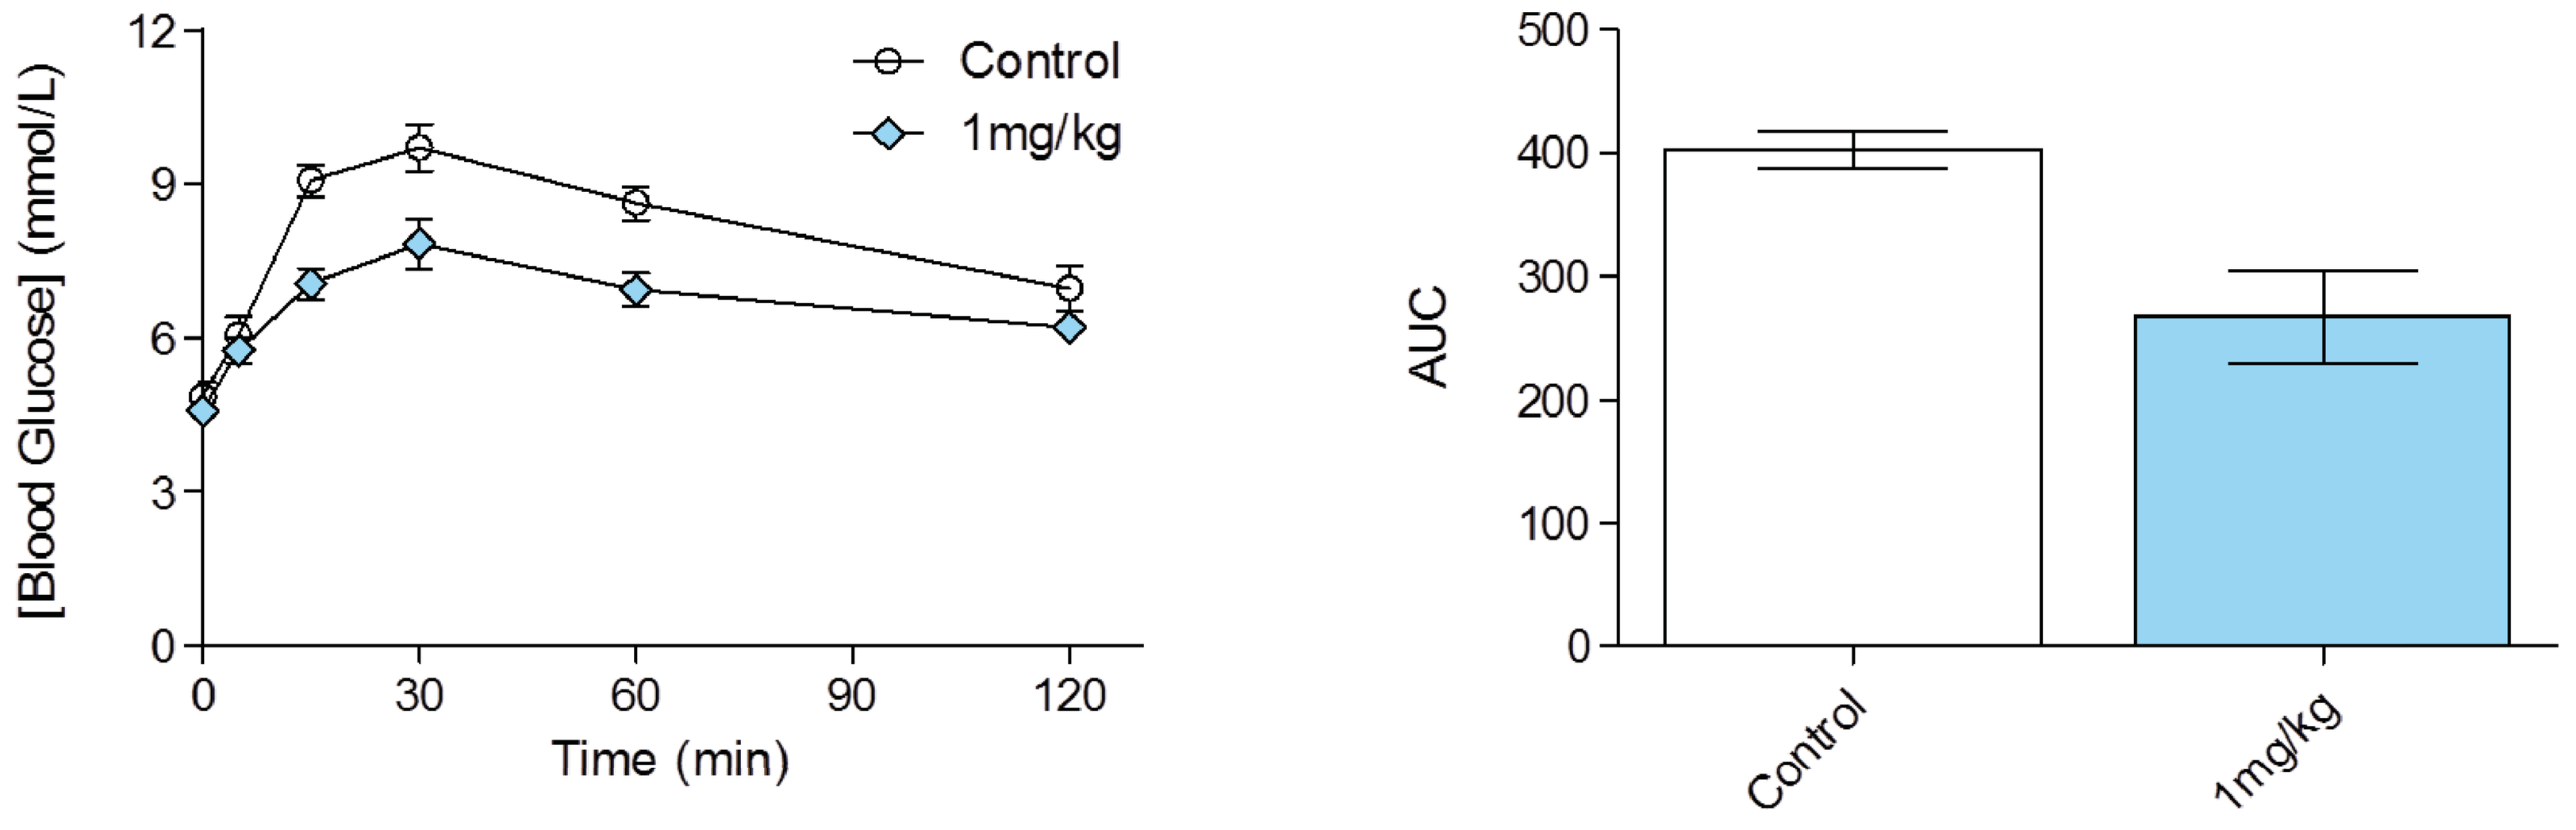
[Blood Glucose] (mmol/L)
Time (min)
Control
1mg/kg
AUC

Figure 2: Blood glucose monitoring in SD rats following oral glucose challenge and AUC

SBM-TFC-039 significantly and rapidly reduces the blood glucose level in diabetic rats. Six (6) hours after the treatment, the glycaemia of SGLT2 inhibitor-treated rats was reduced at the blood glucose level of control fa/+ rats (Figure 3). When challenged with glucose, SBM-TFC-039 at 1mg/kg reduced blood glucose excursion by 89% in ZDF rats.

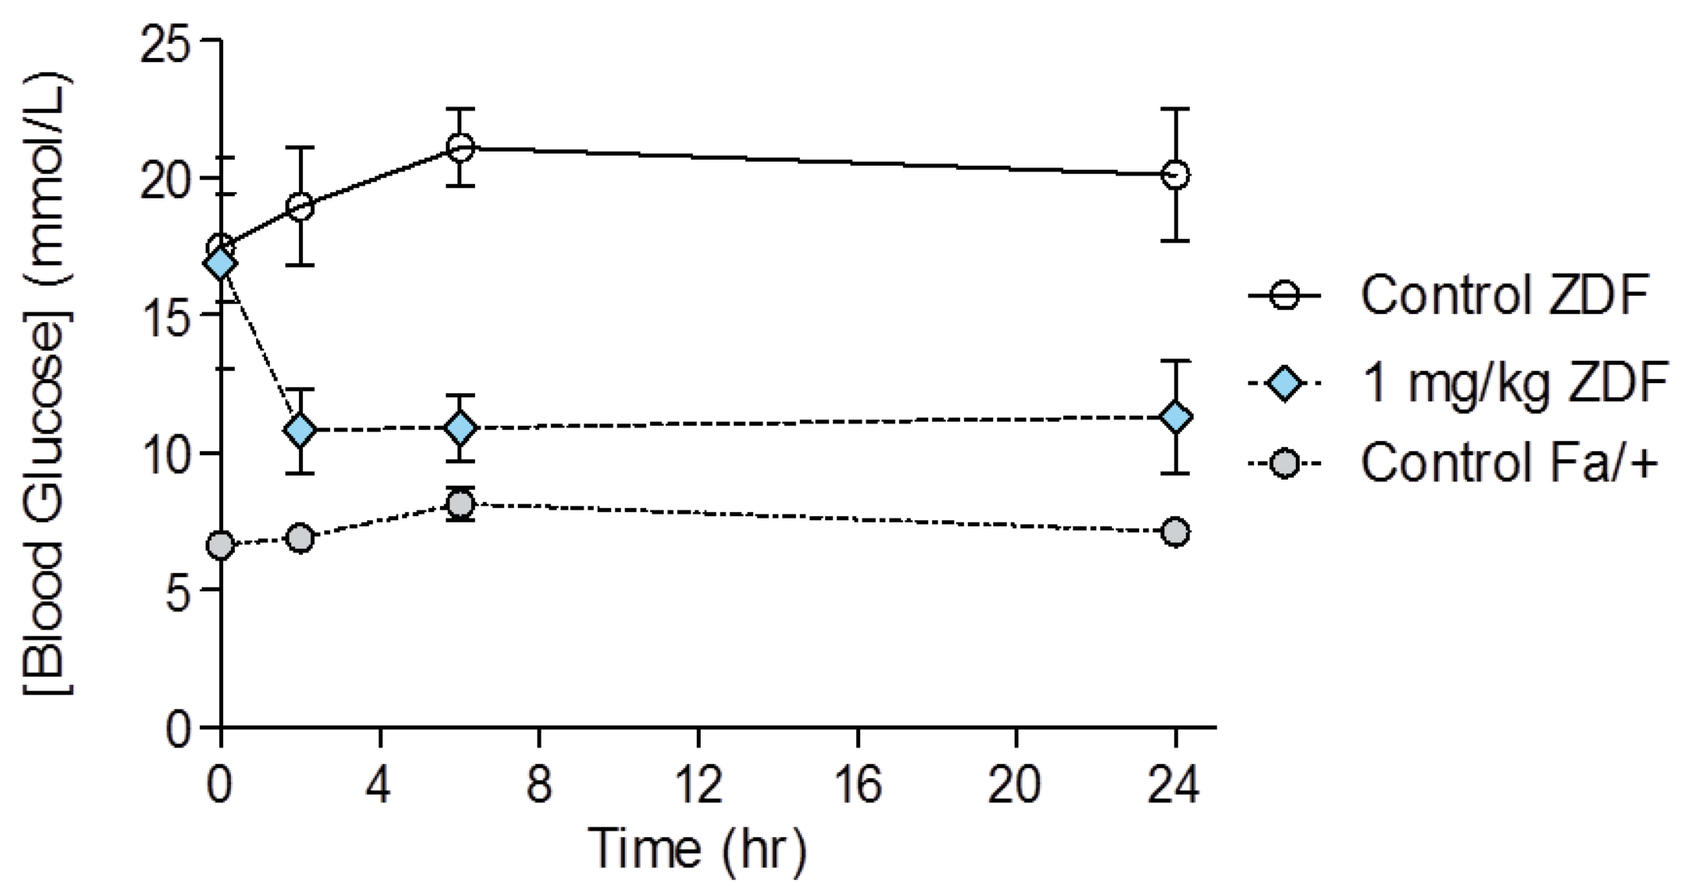
[Blood Glucose] (mmol/L)
Time (hr)
Control ZDF
1 mg/kg ZDF
Control Fa/+

Figure 3: Glycaemia (mmol/L) at 0, 2, 6 and 24 hours after oral administration of SBM-TFC-039 in ZDF rats

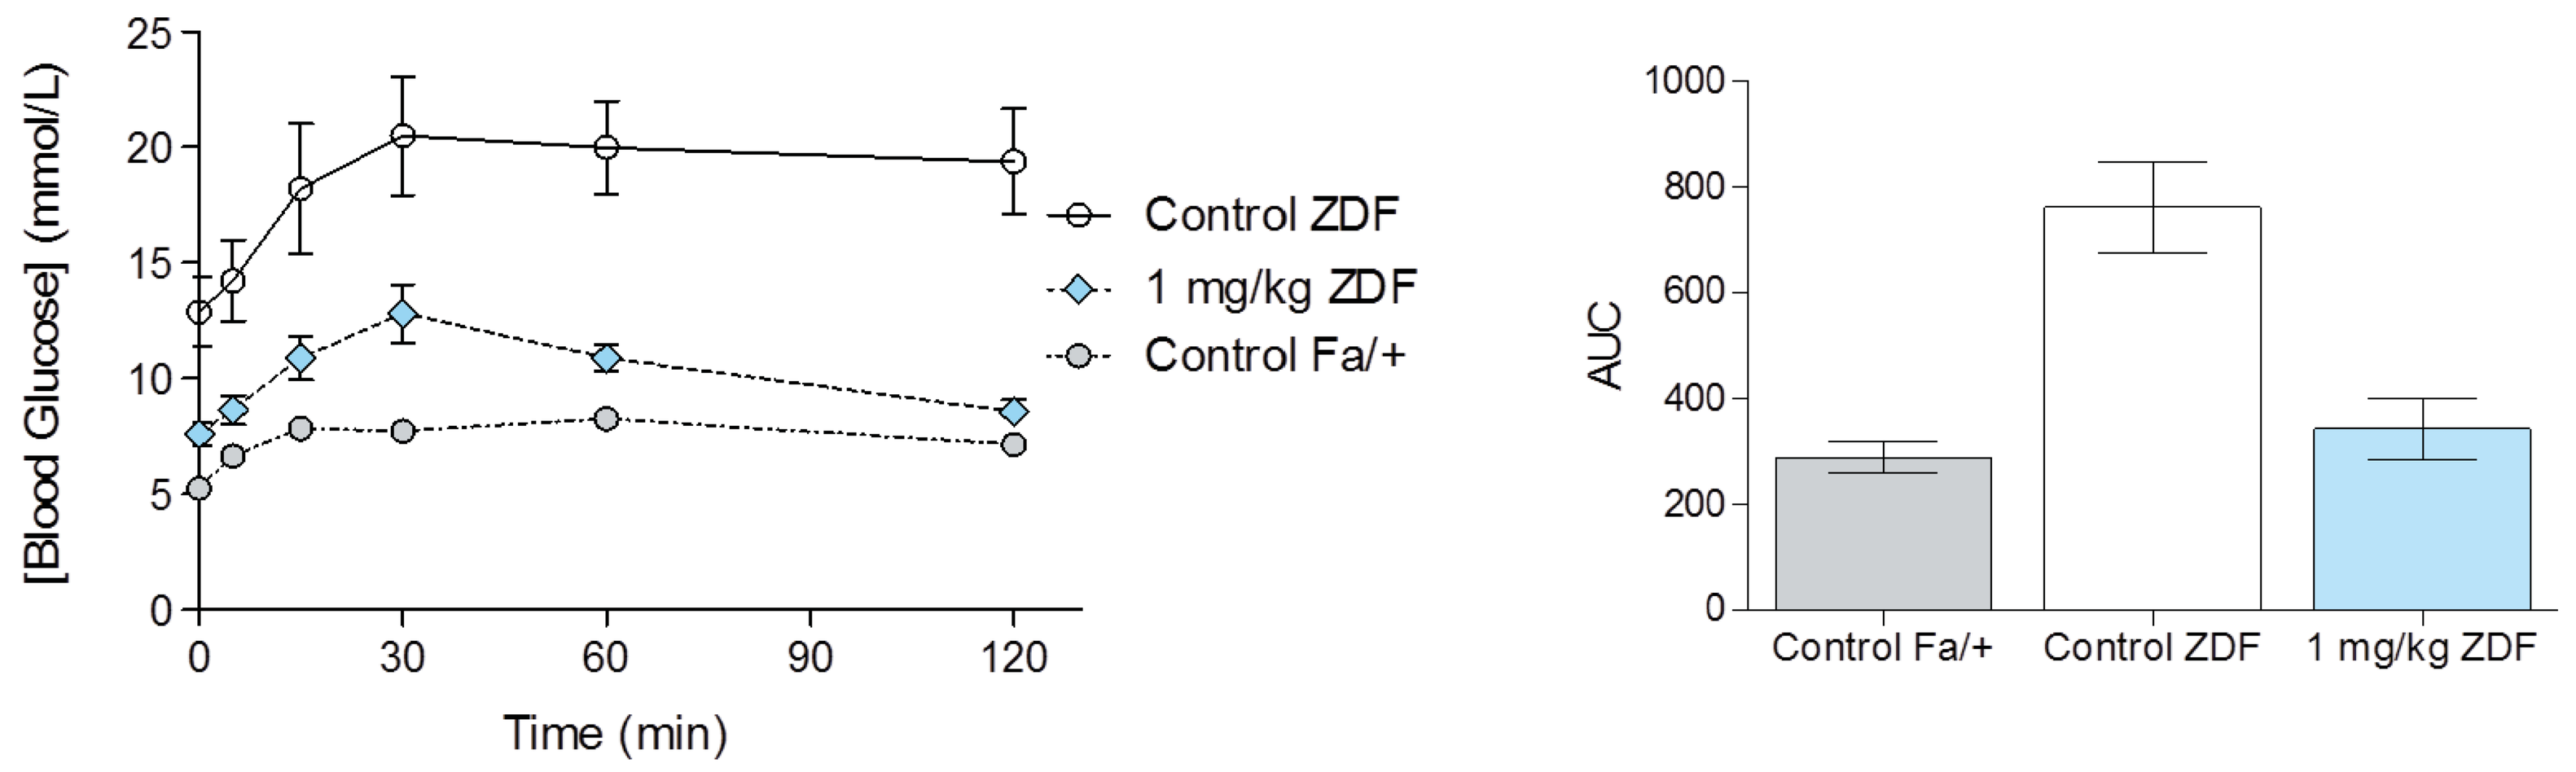
[Blood Glucose] (mmol/L)
Time (min)
Control ZDF
1 mg/kg ZDF
Control Fa/+
AUC
Control Fa/+
Control ZDF
1 mg/kg ZDF

Figure 4: Blood glucose monitoring in ZDF rats following oral glucose challenge and AUC

28-days treatment with SBM-TFC-039 reduced by 71% blood glucose levels in ZDF rats (fed state) compared to non-treated rats (Figure 5, left panel). Plasma fructosamine level in ZDF rats treated for 28 days with SBM-TFC-039 (1 mg/kg) is significantly reduced (65%) compared to control ZDF rats.

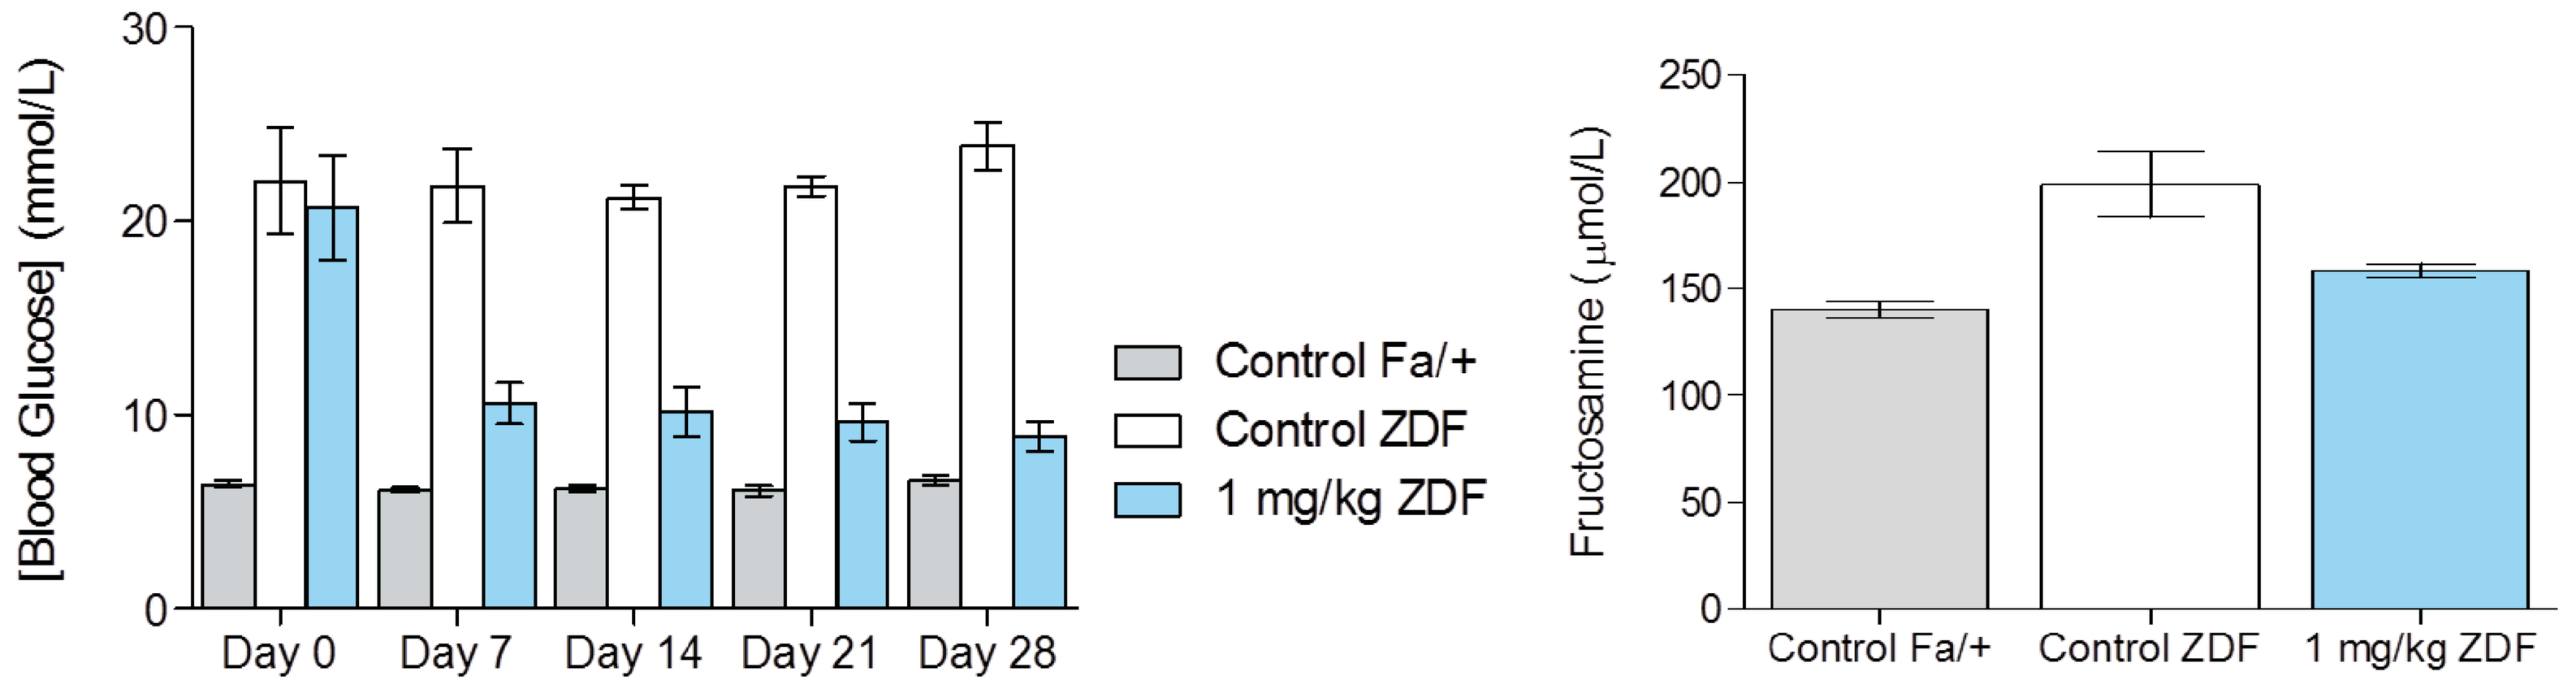
[Blood Glucose] (mmol/L)
Day 0
Day 7
Day 14
Day 21
Day 28
Control Fa/+
Control ZDF
1 mg/kg ZDF
Fructosamine (μmol/L)

Figure 5: Glycaemia (mmol/L) in the fed state at day 0, 7, 14, 21 and 28 (left panel) and fructosamine level following 28 days sub-chronic dosing (right panel)

Conclusion

- SBM-TFC-039 is a novel orally active SGLT inhibitor. SBM-TFC-039 triggers glucosuria and decreases blood glucose excursions following a glucose challenge
- SBM-TFC-039 reduces glycaemia in ZDF rats and in a 28-day chronic dosing study, SBM-TFC-039 normalized diabetes
- Sirona Biochem and TFChem are looking to further characterize their novel SGLT inhibitor

www.sironabiochem.com www.tfchemistry.com